PROTECT™, a Novel Antibody Platform for Integrating Tumor-Specific Immune Modulation and Enhancing the Therapeutic Window of Targeted Multispecific Biologics

Abstract: 924

Florian Heinkel, Anna Von Rossum, Harsh Pratap, Sifa Arrafi, Javairia Rahim, Desmond Lau, Purva Bhojane, Meghan Verstraete, Liz Stangle, Mariam Eji-Lasisi, Veronica Luu, Patrick Farber, Leisa Stenberg, Gesa Volkers, Brandon Clavette, Thomas Spreter von Kreudenstein, Eric Escobar-Cabrera, Surjit Dixit

Zymeworks Inc., 1385 West 8th Ave, Vancouver, BC, Canada

PROTECT™ Platform

PROgrammed **T**umor **E**ngagement & **C**heckpoint/Co-stimulation **T**argeting

Multi-Functional Antibody Design For Tumor-Specific Immune Engagement

- Functional, natural heterodimers (e.g. PD-1/PD-L1) are introduced to sterically block antigen binding outside the tumor. Once at the tumor site, proteases cleave the mask and restore function
- Steric paratope-masking approach is transferable with minimal engineering required to apply to different antibodies
- Use of immunomodulatory domains as masking moieties provides additional functionality that is intrinsic to the mask after tumor specific activation
- Other B7/CD28 family immunomodulatory pairs can provide broad alternative approaches to target specific biologies (e.g. CTLA-4/CD80, CD28/CD80, PDL-1/CD80, ICOS/ICOSL, CD47/SIRPa)

Off-tumor

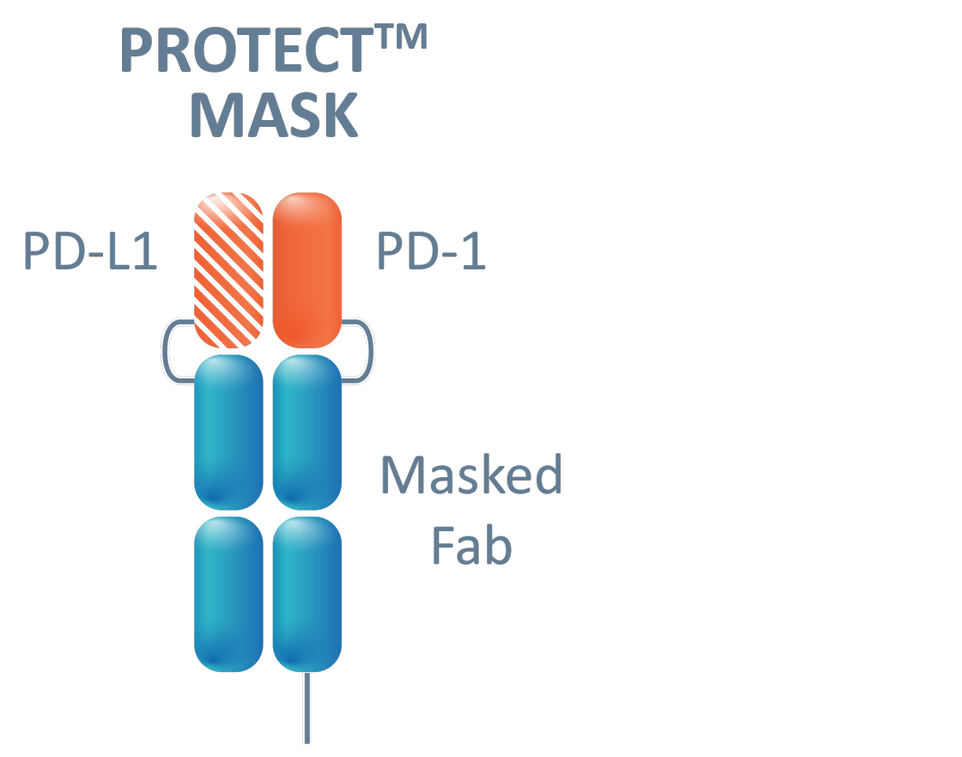
PROTECT™
MASK
PD-L1
PD-1
Masked
Fab

- Antibody activity is masked to enhance tolerability by avoiding on-target, off-tumor toxicities
- Target mediated drug disposition is avoided
- Therapeutic window increased by reducing toxicity

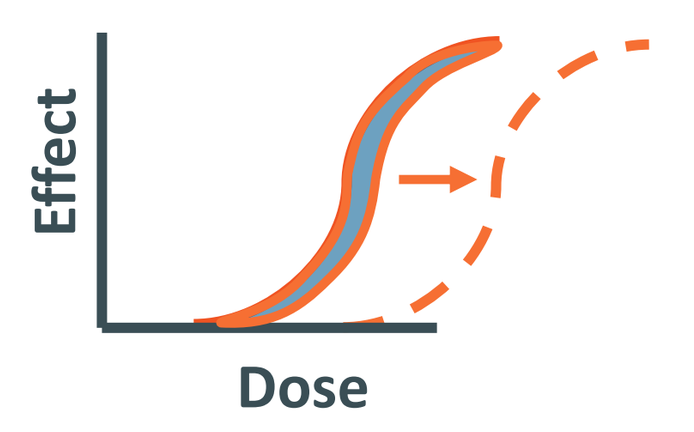
Effect
Dose

Tumor

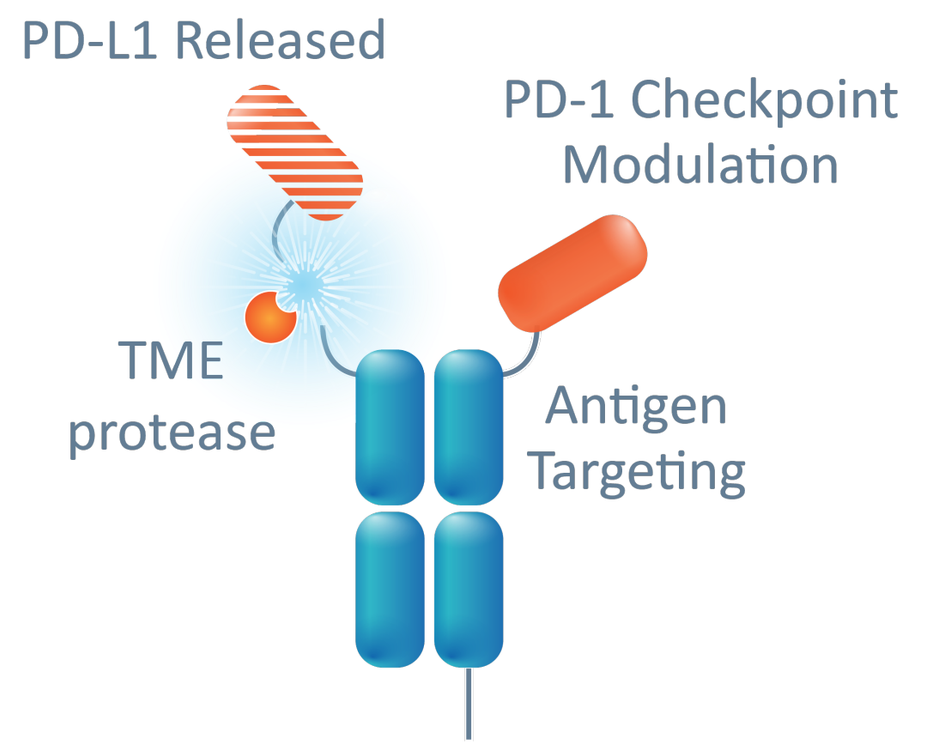
PD-L1 Released
PD-1 Checkpoint
Modulation
TME
protease
Antigen
Targeting

- Tumor-specific proteases cleave and release the steric mask to activate the biologic
- The fusion B7 domain targets an immune checkpoint in concert with antigen binding to achieve bispecific engagement
- Therapeutic window increased by enhancing potency

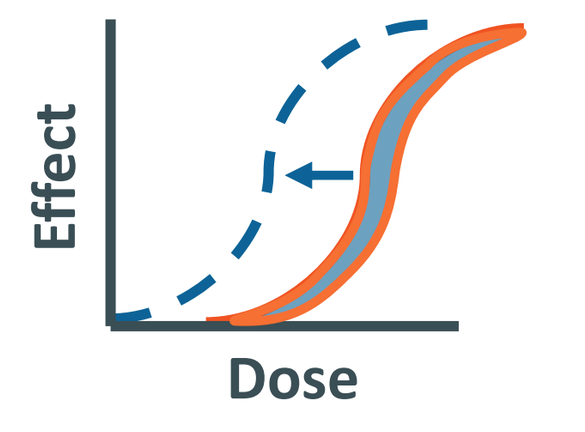
Effect
Dose

Evolution of CD3-Redirecting Bispecifics

Traditional CD3 Approach

- 1:1 targeting of TAA and CD3
- Affinity/valency engineering to reduce required CD3 potency
- Fc fusions improve PK and CMC

Challenges
- Systemic CRS toxicities
- Risk of on-target off-tumor toxicities
- Limited utility in solid tumor setting

Current Masked CD3 Approach

- Customized tumor-specific activity via conditional masking to reduce off-tumor toxicities

Challenges:
- Solid tumor applications yet to be clinically validated
- Need to tailor each mask on a per program basis
- Cold immune exhausted phenotype, checkpoint regulation limiting activity

Next-Gen PROTECT-CD3™

- **Plug-and-play, transferable** tumor-specific activity via conditional, steric masking to reduce off-tumor toxicities
- **Avidity** in target engagement with multispecific binding
- **Functional** mask adds **co-stimulation/checkpoint blockade** to enhance efficacy in the tumor
- Built on the foundation of the **Azymetric™ bispecific platform**
- **Unique geometry** for T-cell bridging

PROTECT-CD3™:

Conditionally-Active T-Cell Engaging Trispecific

Architecture of PROTECT-CD3™

- PD-1 / PD-L1 heterodimer is leveraged as the masking module for an αCD3 bispecific
- An engineered protease cleavage site is introduced to release the PD-L1 domain in the TME
- Azymetric™ platform was used to generate bispecific antibody
- EFECT™ platform was used to reduce Fc effector function

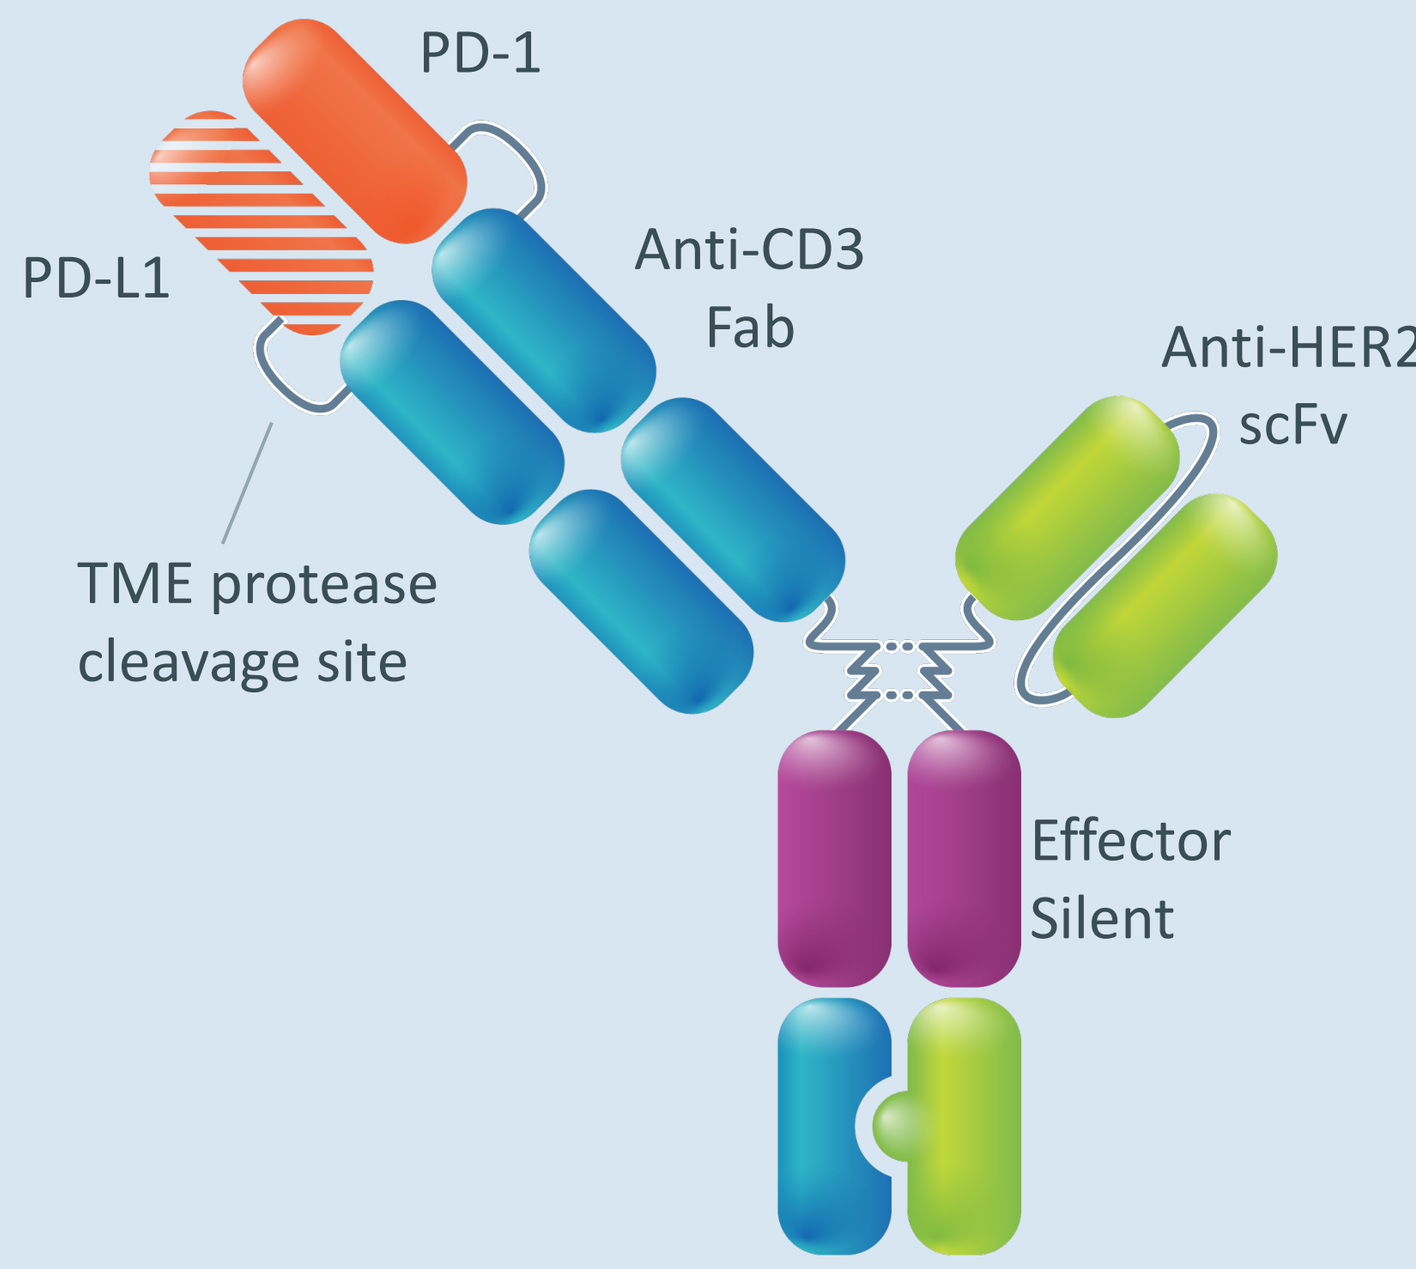
PD-1
PD-L1
Anti-CD3
Fab
Anti-HER2
scFv
TME protease
cleavage site
Effector
Silent

Proposed Mechanism of Action of PROTECT-CD3™

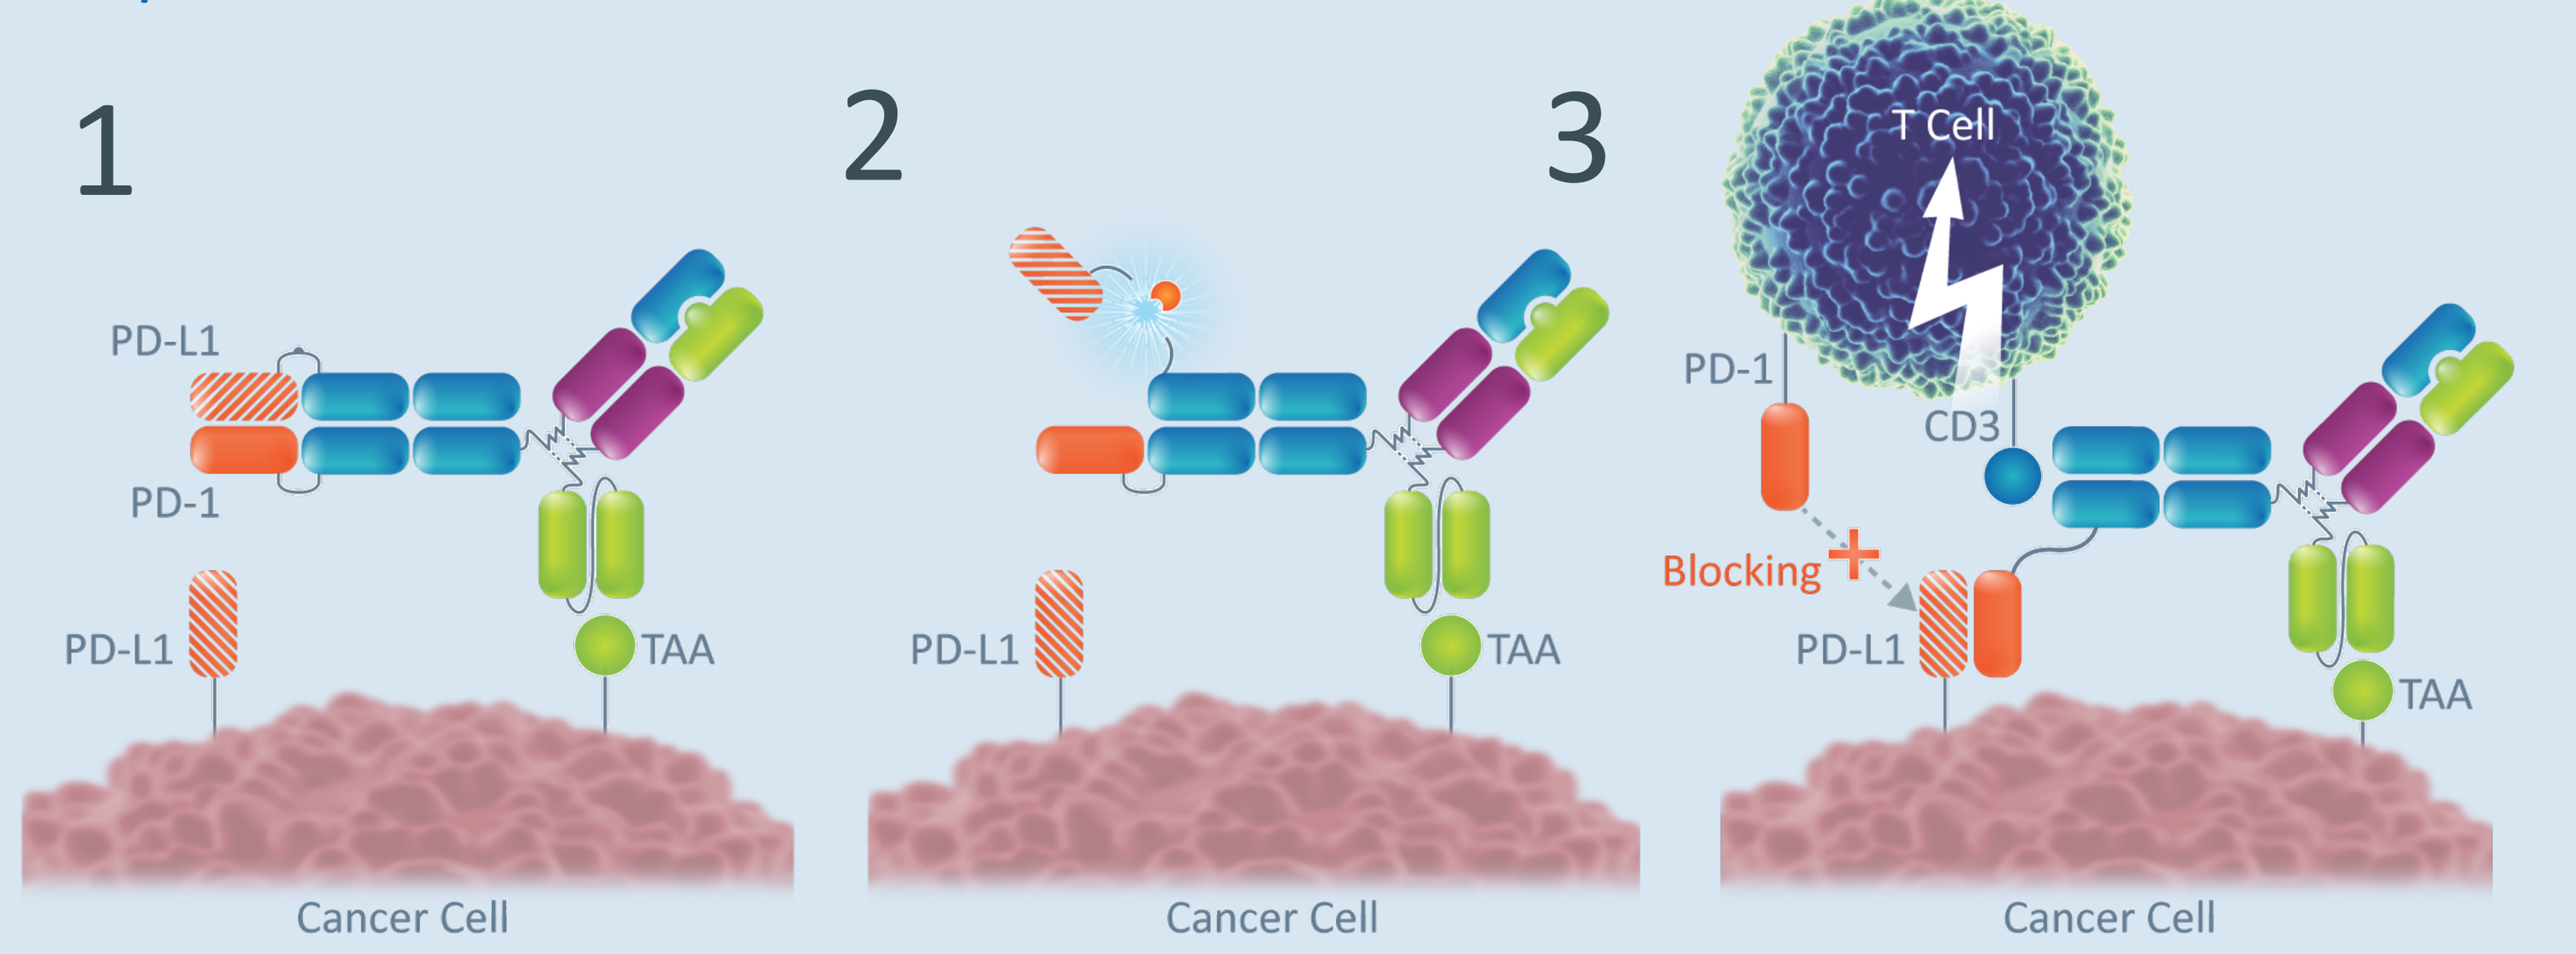
1
2
3
T Cell
PD-L1
PD-1
TAA
CD3
Blocking
Cancer Cell

1. TAA arm (e.g. αHER2) directs PROTECT-CD3™ therapeutic to TME
2. TME protease releases PD-L1 moiety of the mask
3. Activated PROTECT-CD3™ trispecific binds to TAA, PD-L1 and CD3

PD-1/PD-L1 is an Effective Mask for CD3 Binding; Cleavage of Mask Restores Binding to CD3+ cells

- Binding to CD3+ (Pan T) cells was determined by flow cytometry
- Binding of high-affinity αCD3 paratope was confirmed (EC50 = 2.3 nM)
- Masking effectively reduced binding to CD3 by > 100-fold
- Upon protease-based activation, CD3 binding recovery was observed

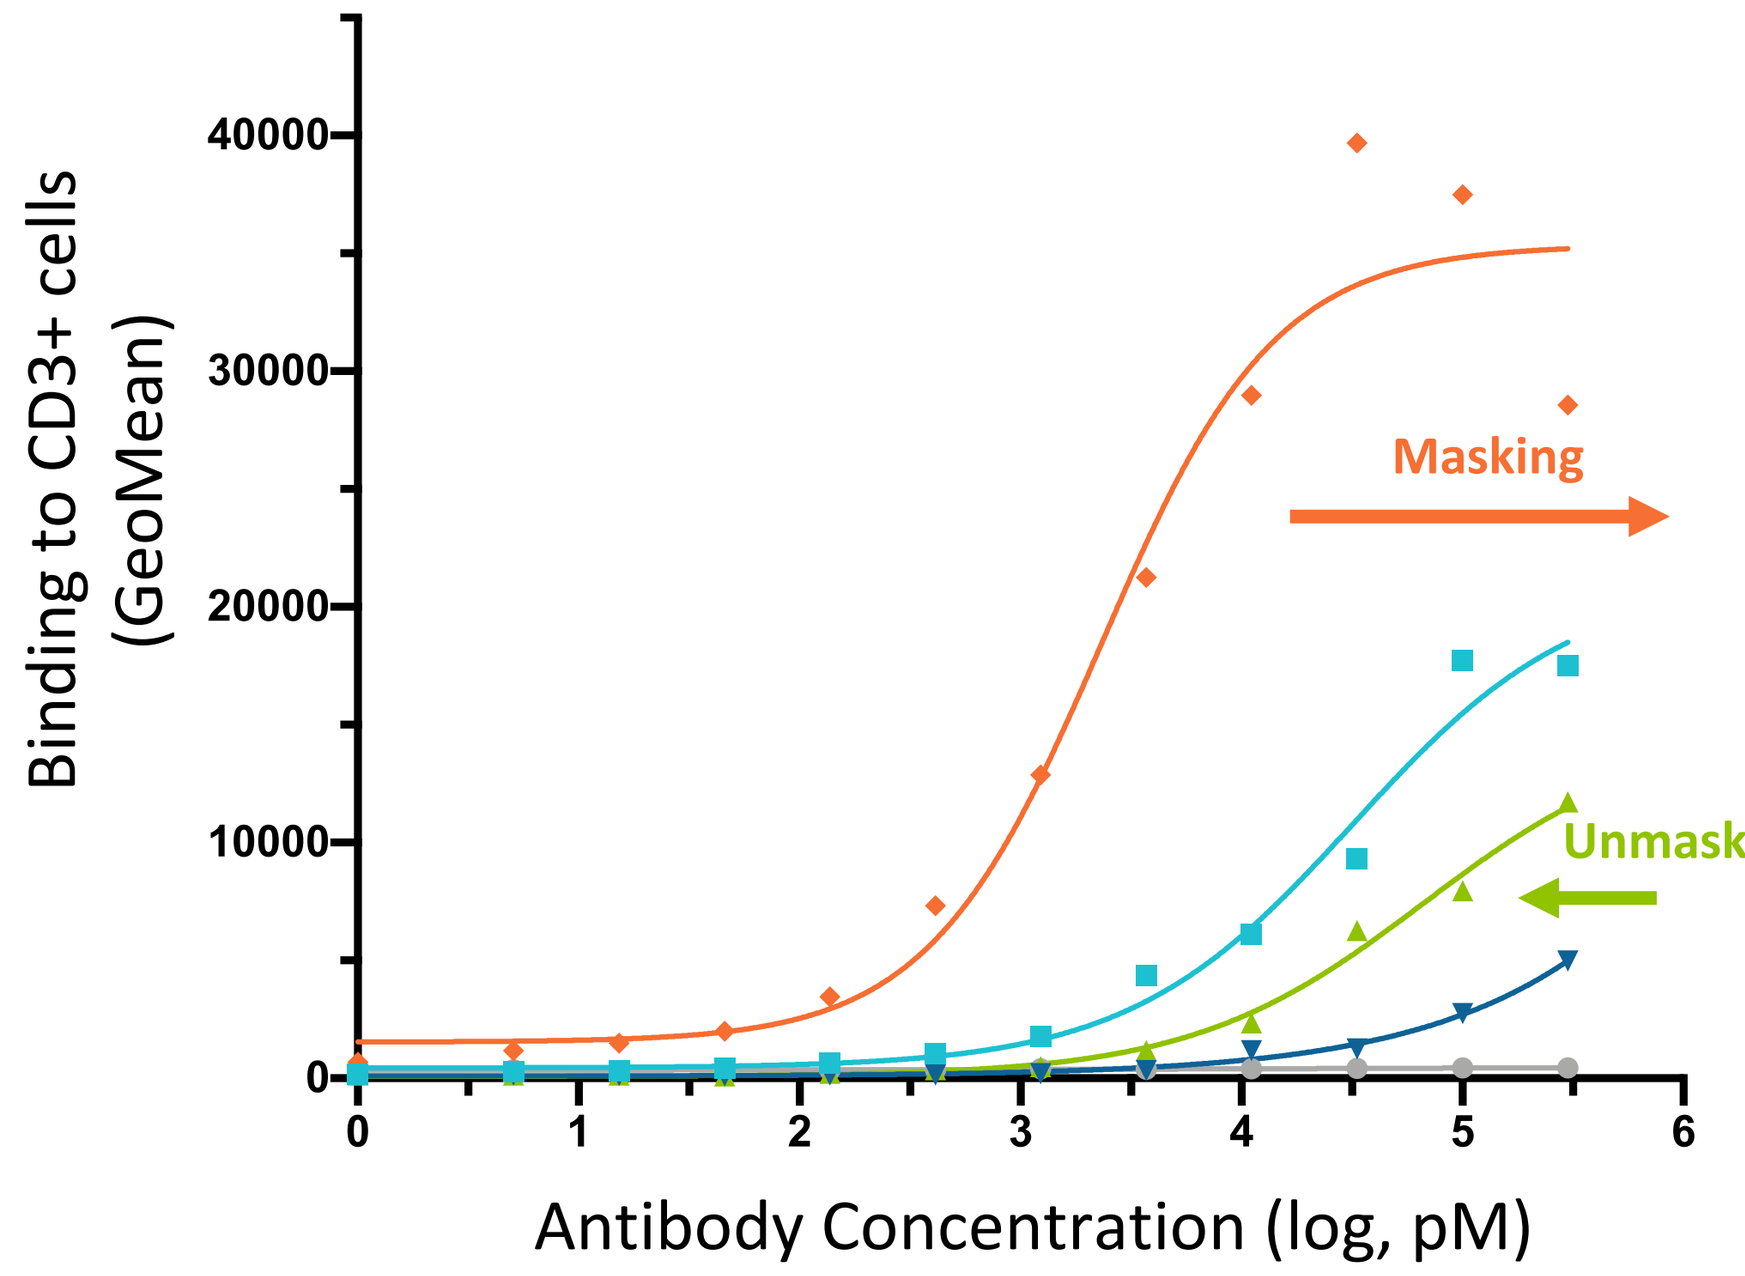
Binding to CD3+ cells
(GeoMean)
40000
30000
20000
10000
0
0
1
2
3
4
5
6
Masking
Unmasking
Antibody Concentration (log, pM)

Description	Cartoon/Legend
+ve Bispecific Control (CD3-HER2)	
Trispecific Control (PD-L1-CD3-HER2)	
Masked PROTECT-CD3™ Trispecific (PD-1/PD-L1 Masked CD3-HER2)	
Unmasked PROTECT-CD3™ Trispecific (PD-L1-CD3-HER2)	
Irrelevant –ve control	

PD-1/PD-L1 Pair is an Effective Mask for PD-L1 Binding; Cleavage of Mask Restores Binding to PD-L1

- Binding to PD-L1-transfected CHO-cells was measured by flow cytometry
- Trispecific control shows similar affinity to PD-1 Fc
- > 100-fold reduced binding to PD-L1 by masking
- Upon protease-based activation, PD-L1 binding recovery was detected

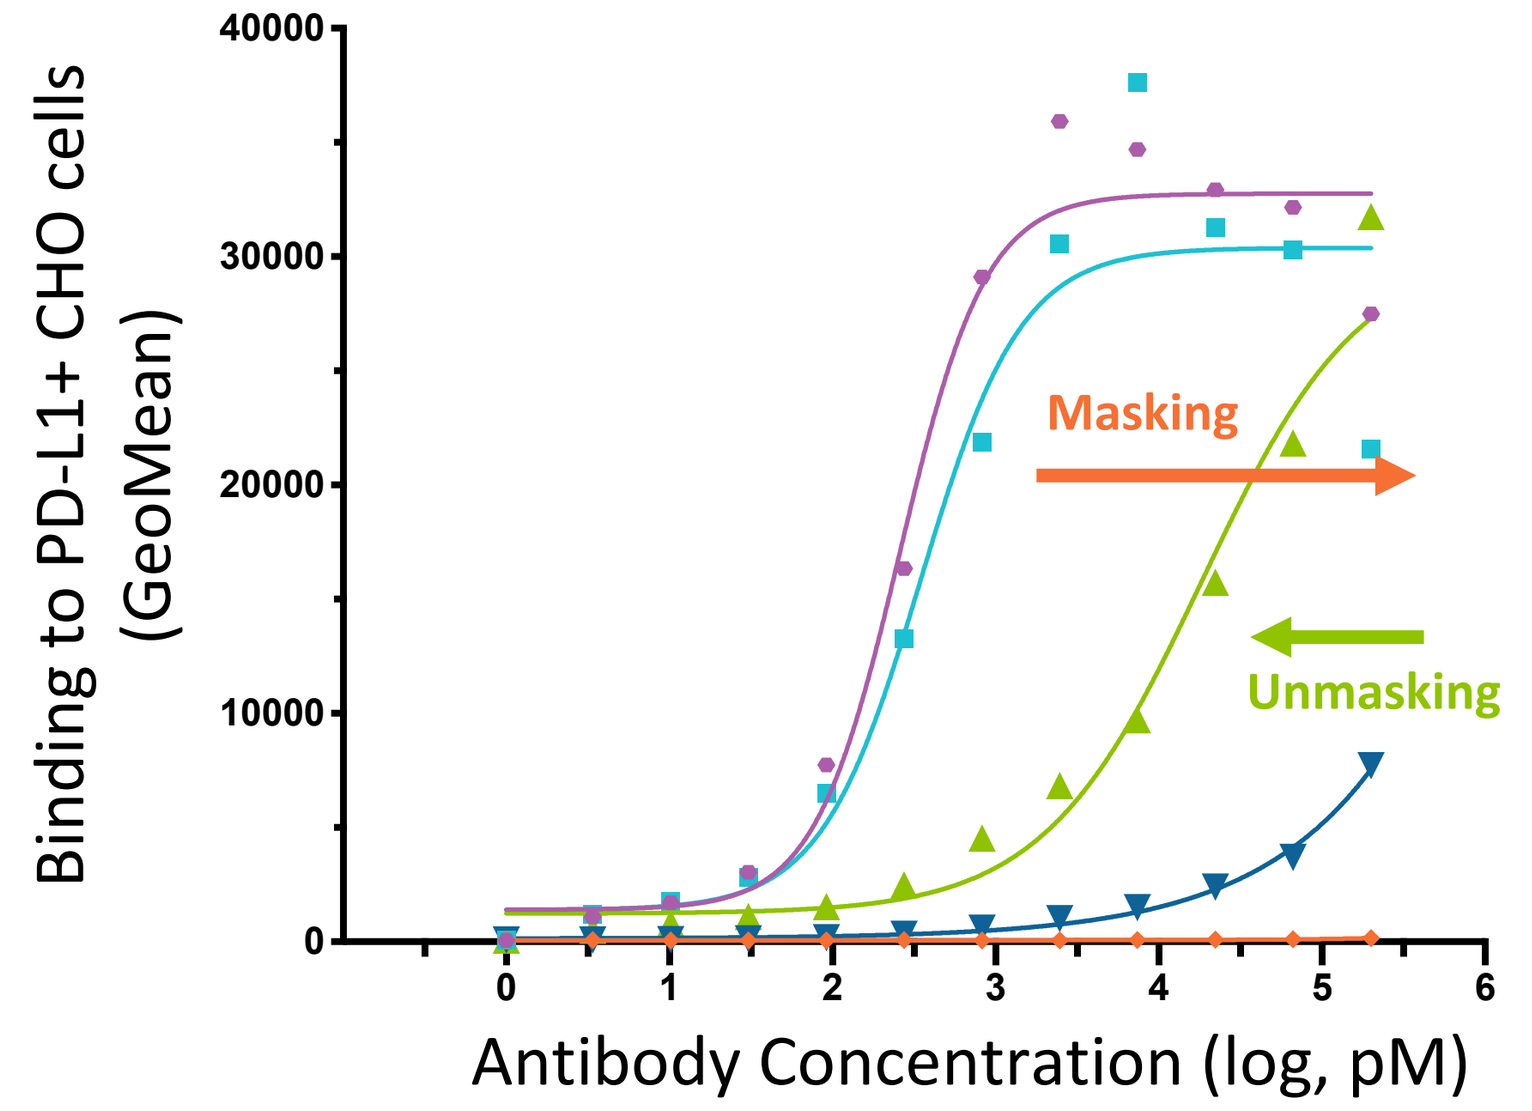
Binding to PD-L1+ CHO cells
(GeoMean)
40000
30000
20000
10000
0
0
1
2
3
4
5
6
Masking
Unmasking
Antibody Concentration (log, pM)

Description	Cartoon/Legend
PD-1 Fc	
Trispecific Control (PD-L1-CD3-HER2)	
Masked PROTECT-CD3™ Trispecific (PD-1/PD-L1 Masked CD3-HER2)	
Unmasked PROTECT-CD3™ Trispecific (PD-L1-CD3-HER2)	
Bispecific Control (CD3-HER2)	

PROTECT™ Mask Significantly Reduces T-Cell Dependent Cellular Cytotoxicity; Enhanced Cytotoxicity Observed with PROTECT-CD3™ after Protease Activation

- T-cell dependent cytotoxicity was determined in co-culture assay of an endogenous HER2+/PD-L1+ cancer cell line (JIMT-1) with Pan T-cells at a 5:1 effector:target ratio
- 19-fold reduction of TDCC potency was seen for masked variant compared to bispecific control
- 18-fold enhanced potency in TDCC was observed with PD-1 checkpoint modulation after protease activation compared to bispecific control
- Activated trispecific showed increased potency compared to the combination of atezolizumab (αPD-L1) with the bispecific
- Reduction of T-cell cytotoxicity by masking and recovery and increased potency after unmasking was confirmed by IFNɣ readout

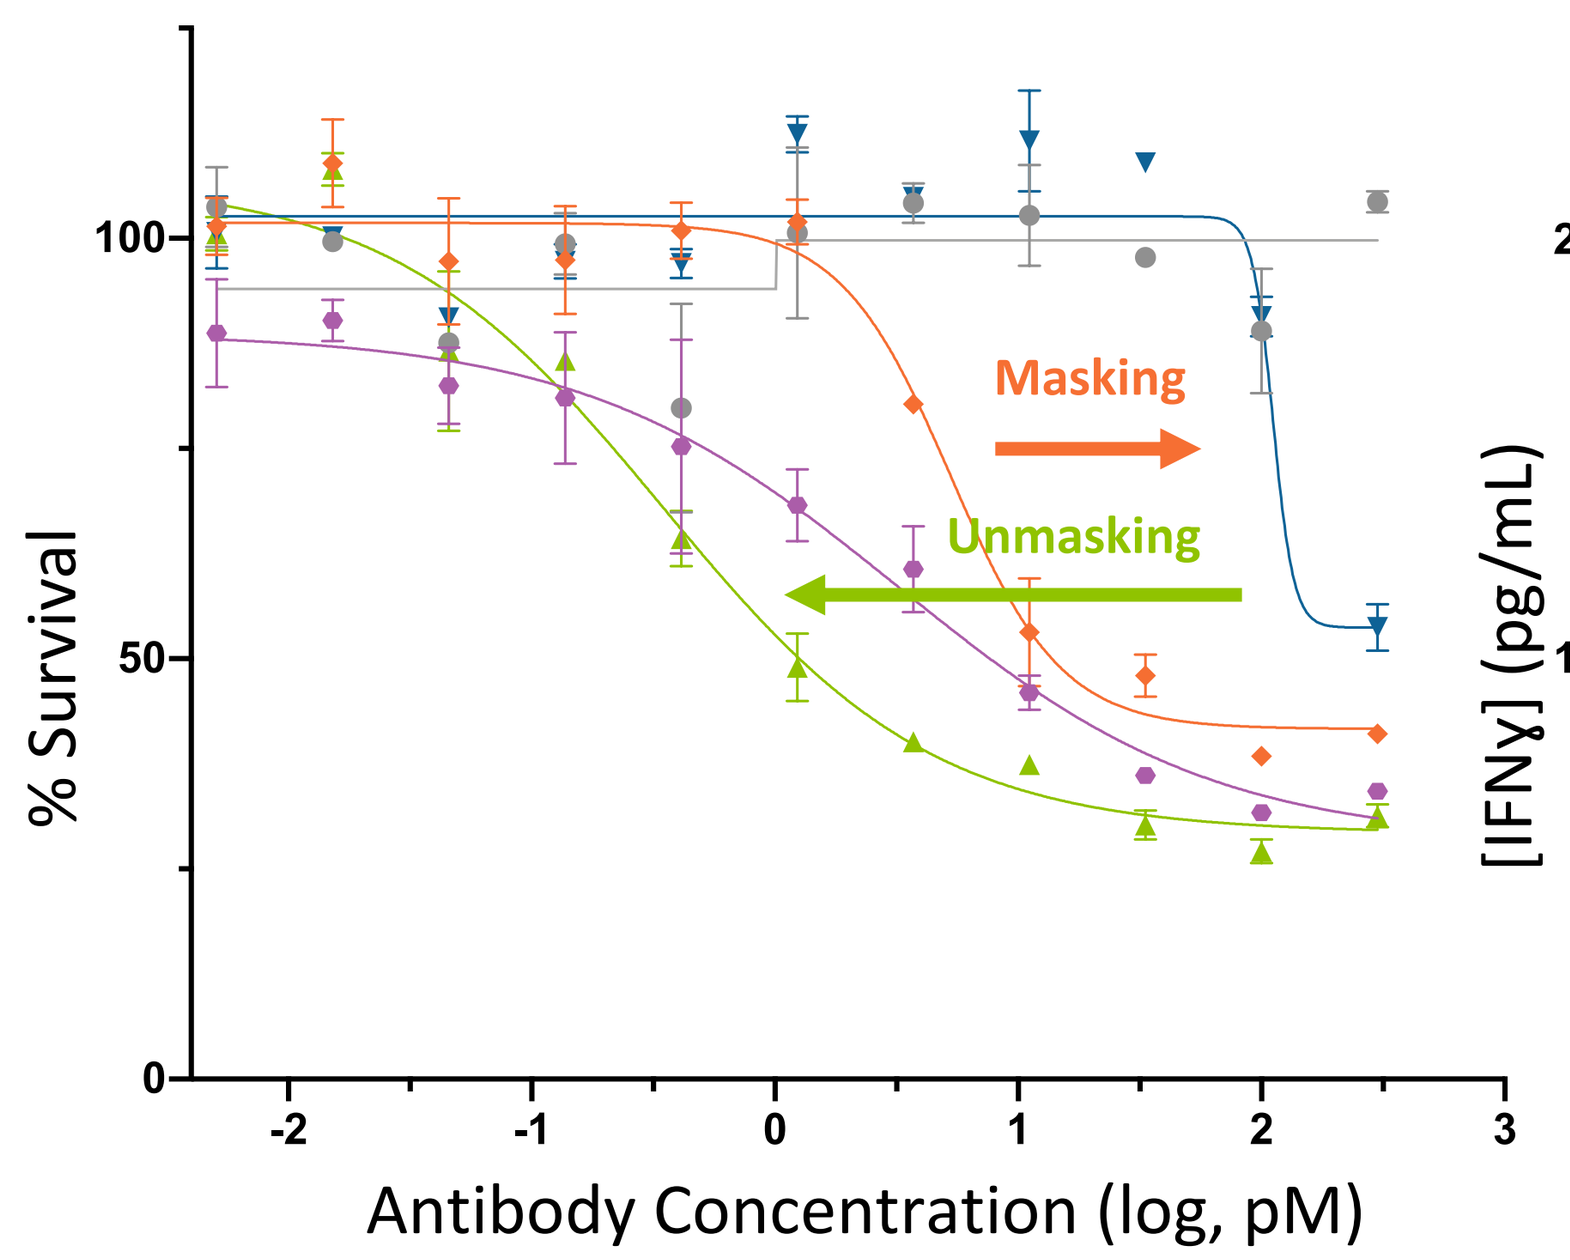
% Survival
100
50
0
-2
-1
0
1
2
3
Masking
Unmasking
Antibody Concentration (log, pM)

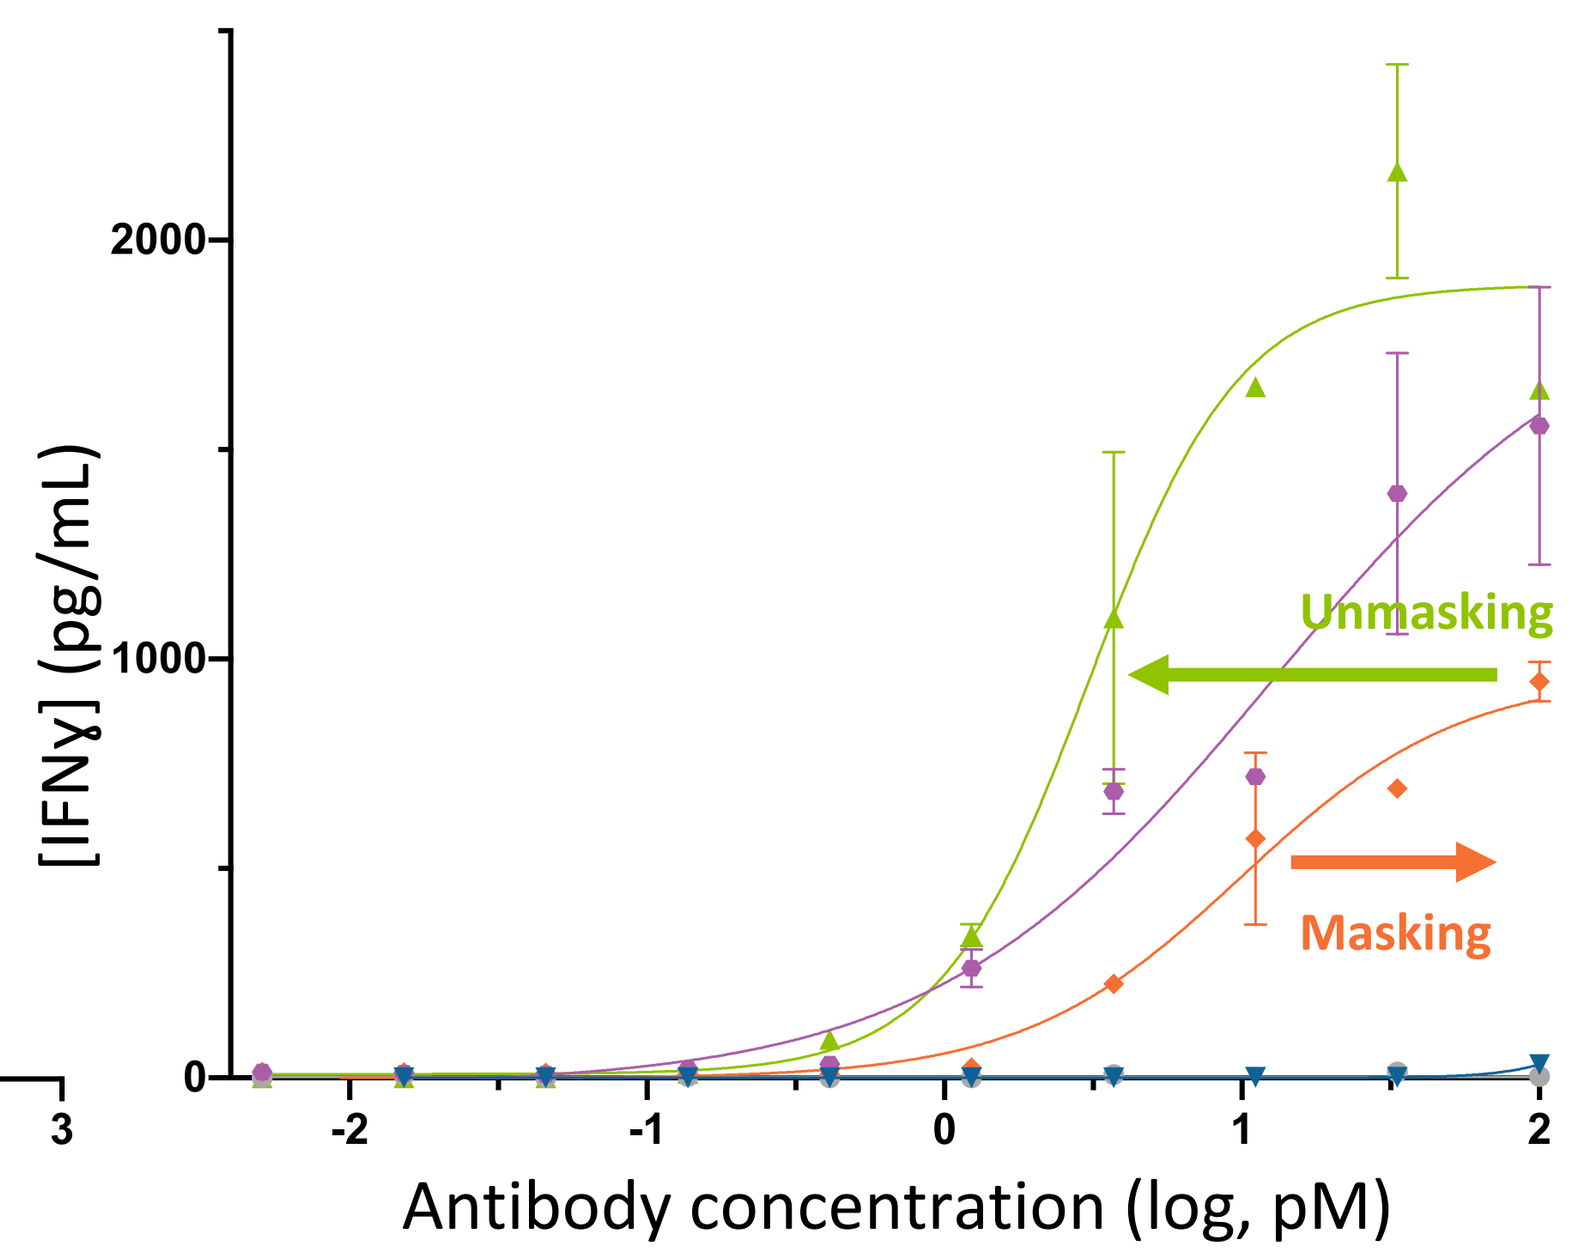
[IFNɣ] (pg/mL)
2000
1000
0
-2
-1
0
1
2
Unmasking
Masking
Antibody concentration (log, pM)

Description	Cartoon/Legend	Survival EC50 (pM)	IFNɣ EC50 (pM)
+ve Bispecific Control (CD3-HER2)		5.3	10
Masked PROTECT-CD3™ Trispecific (PD-1/PD-L1 Masked CD3-HER2)		> 100	N.S*
Unmasked PROTECT-CD3™ Trispecific (PD-L1-CD3-HER2)		0.3	3
+ve Bispecific Control + 120 nM Atezo (αPD-L1)	+	3.2	13
Irrelevant –ve control		N.S.*	N.S.*

*N.S.: No signal detected under the conditions tested

PD-L1, HER2 and CD3 are Simultaneously Engaged by PROTECT-CD3™ Trispecific

- Binding to an endogenous HER2+/PD-L1+ cancer cell line (JIMT-1, receptor ratio of HER2/PD-L1: 3:1) was measured by flow cytometry (1 nM concentration point shown)
- Higher MFI for trispecific (PD-1-CD3-HER2) compared to bispecific controls suggest that both PD-L1 and HER2 are simultaneously engaged on the cancer cell supporting the proposed MOA
- Antibody-dependent bridging of a HER2+/PDL1+ cancer cell line and Pan T-cells was assessed by the presence of signal for double positive signal (fluorescent signal for both T-cell and target cell at the same time) in flow cytometry
- Higher % of bridging T-cells and cancer cells for trispecific (PD-1-CD3-HER2) compared to bispecific controls suggest all three targets are simultaneously engaged

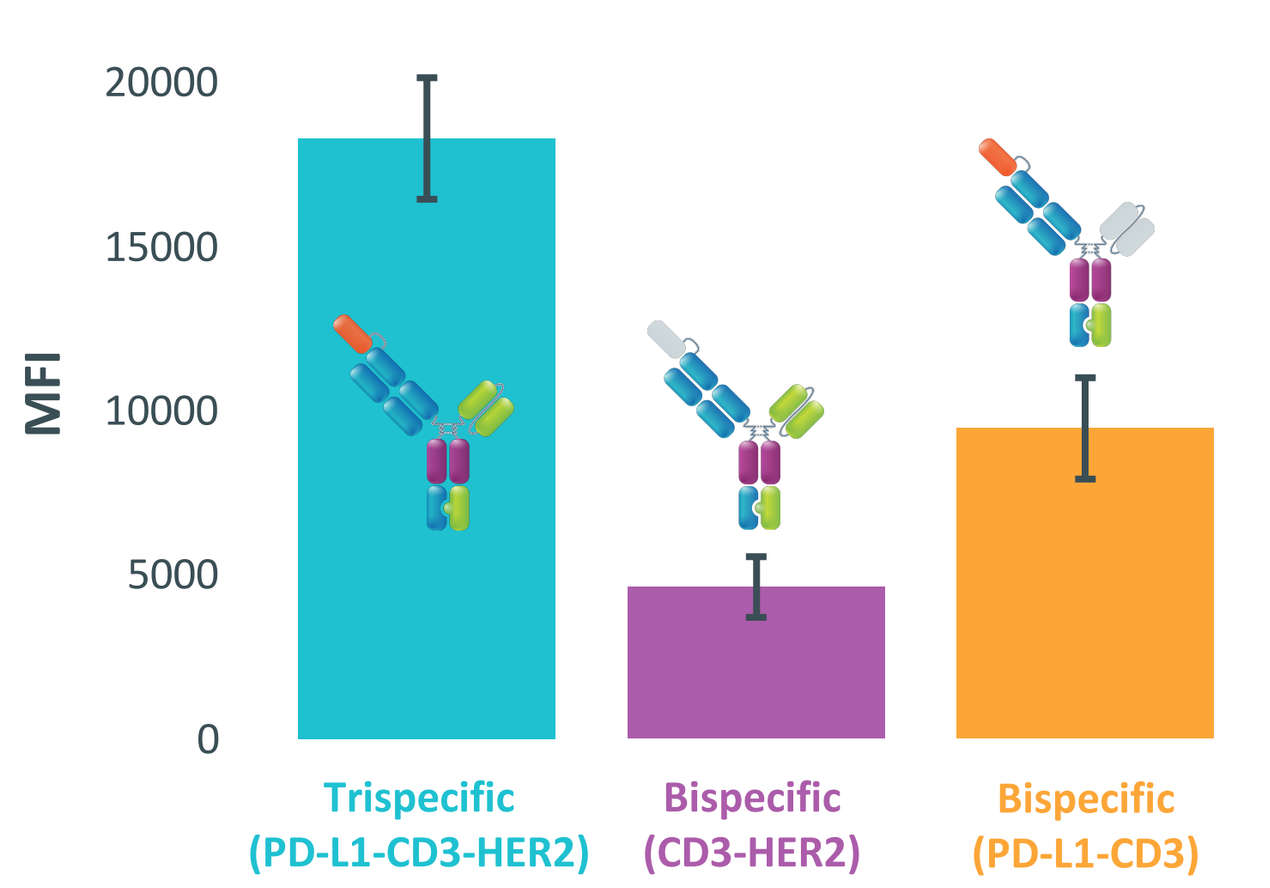
MFI
20000
15000
10000
5000
0
Trispecific
(PD-L1-CD3-HER2)
Bispecific
(CD3-HER2)
Bispecific
(PD-L1-CD3)

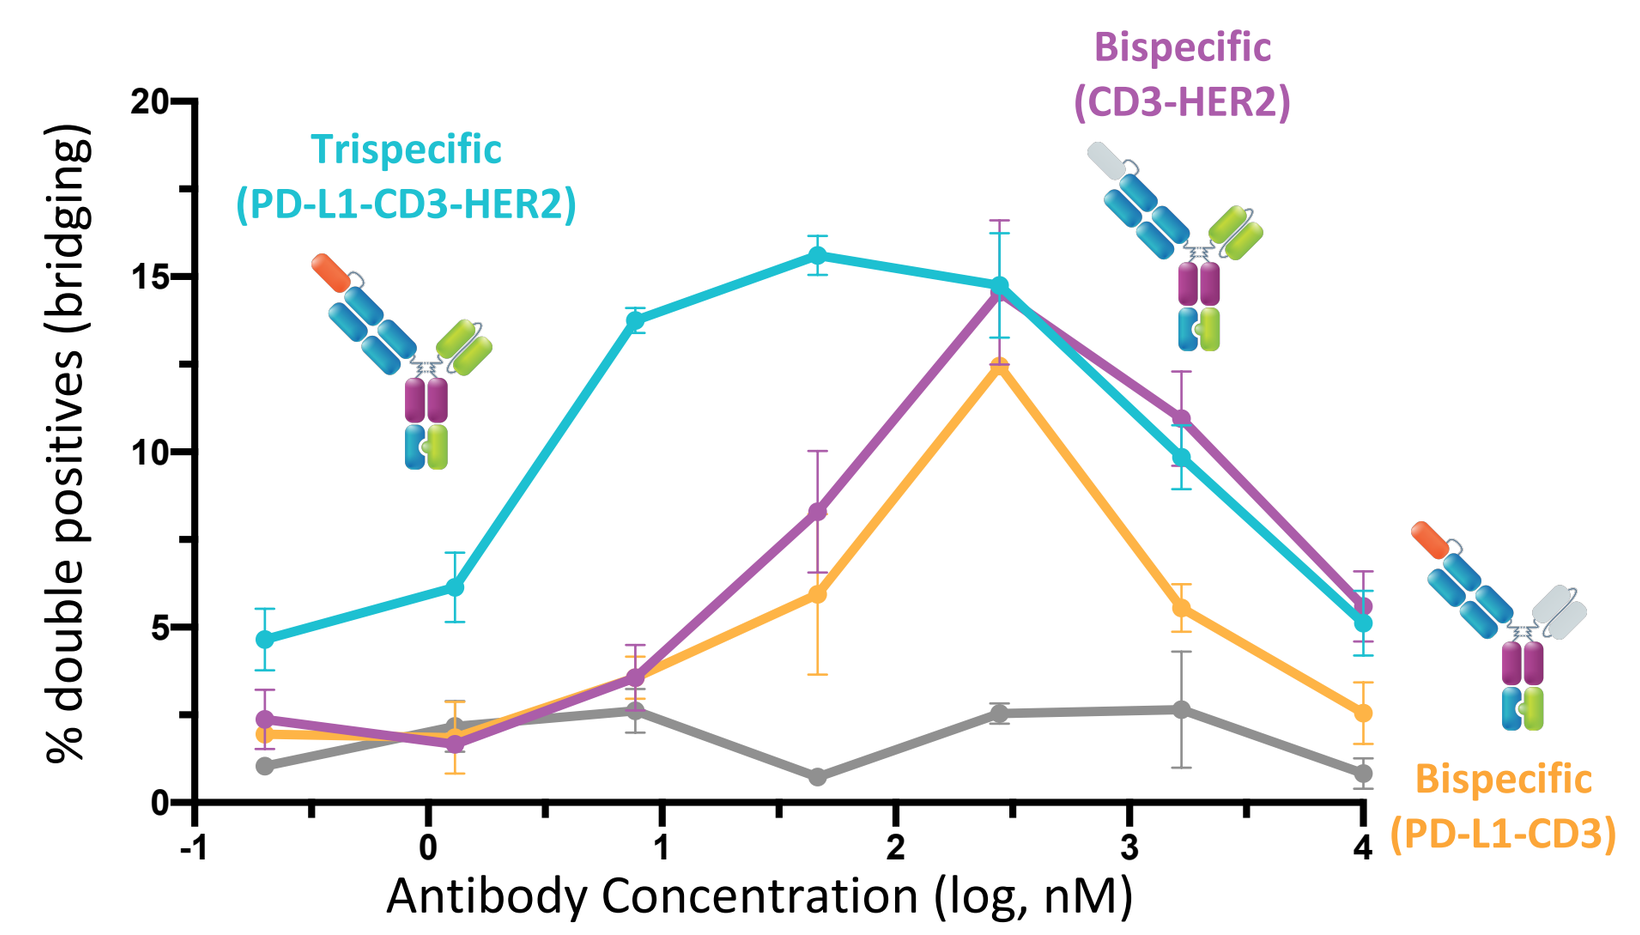
% double positives (bridging)
20
15
10
5
0
-1
0
1
2
3
4
Trispecific
(PD-L1-CD3-HER2)
Bispecific
(CD3-HER2)
Bispecific
(PD-L1-CD3)
Antibody Concentration (log, nM)

Universal Application of PROTECT™

PROTECT™ PD-1/PD-L1 Based Mask is Transferable to Other Target Paratopes as Determined by Masking & Recovery of Target Binding

- 10-1000-fold decrease in antigen binding by masking
- Partial recovery of binding upon cleavage was seen for systems with cleavable mask

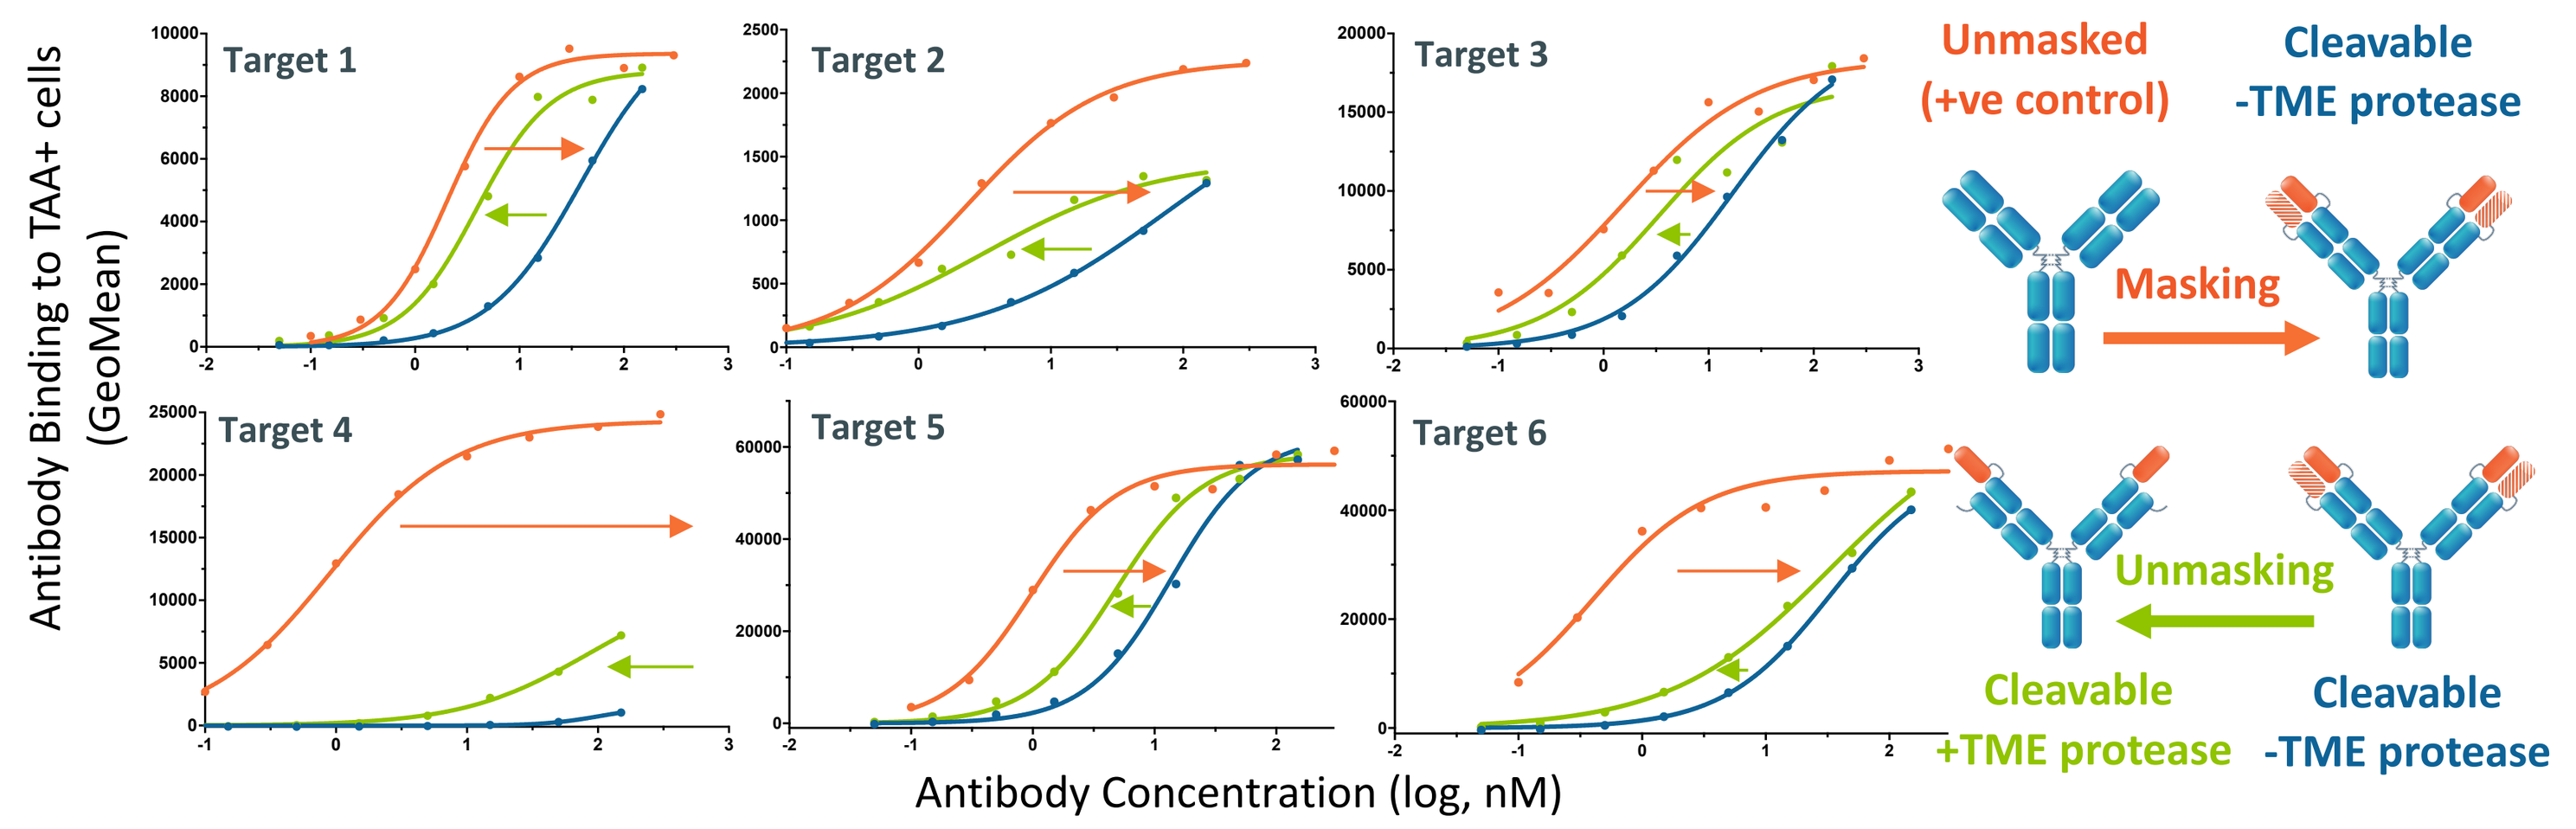
Antibody Binding to TAA+ cells
(GeoMean)
Target 1
Target 2
Target 3
Target 4
Target 5
Target 6
Unmasked
(+ve control)
Cleavable
-TME protease
Masking
Unmasking
Cleavable
+TME protease
Cleavable
-TME protease
Antibody Concentration (log, nM)

Conclusion

- Novel first-in-class approach to combine transferable, tumor-specific masking/unmasking approach with immune modulation to enhance the therapeutic window for broad classes of therapeutics
- Demonstrated potential to enable better tolerated and more potent next-generation CD3-engaging multispecifics
- Demonstrated application of steric masking/unmasking approach to variety of paratopes with minimal engineering required
- Zymeworks is progressing with the development of therapeutic PROTECT-CD3™ programs

Acknowledgements

The work presented was supported by Stephanie Masterman and Kara White Moyes

AACR Annual Meeting 2021